Is HCV elimination among HIV-infected people who inject drugs possible through HCV treatment targeting HIV/HCV coinfection? A modeling analysis for Andalusia, Spain

BRITT SKAATHUN[1], ANNICK BORQUEZ[1], ANTONIO RIVERO-JUAREZ[2], FRANCISCO TELLEZ[3], MANUEL CASTAÑO[4], DOLORES MERINO[5], JESUS SANTOS[6], JUAN MACÍAS[7], A RIVERO[2], NATASHA K MARTIN[1,8]

[1]Division of Infectious Diseases and Global Public Health, University of California San Diego, USA. [2]Infectious Diseases Unit, Instituto Maimonides de Investigaciones Biomedicas de Cordoba (IMIBIC), Hospital Universitario Reina Sofia de Cordoba, Universidad de Cordoba, Spain. [3]Infectious Diseases Unit Hospital Universitario de Puerto Real, Instituto de Investigación e Innovación en Ciencias Biomédicas de la Provincia de Cádiz. Universidad de Cádiz. Cádiz, Spain. [4]Infectious Diseases Unit. Hospital Regional Universitario de Málaga. Málaga, Spain. [5]Infectious Diseases Unit. Hospitales Juan Ramón Jiménez e Infanta Elena de Huelva. Huelva, Spain. [6]Infectious Diseases Unit. Hospital Universitario Virgen de la Victoria. Complejo Hospitalario Provincial de Málaga. Málaga, Spain. [7]Unidad de Enfermedades Infecciosas. Hospital Universitario de Valme. Instituto de Biomedicina de Sevilla (iBiS). Sevilla, Spain. [8]School of Social and Community Medicine, University of Bristol, UK.

Introduction

- Scale-up of HCV treatment for HIV/HCV coinfected individuals is occurring in Spain, the majority with a history of injecting drug use (IDU).[1]
- It is unclear if this scale-up will achieve the WHO elimination target (90% reduction in incidence by 2030) among HIV-infected people who inject drugs (HIV+ PWID).[2]
- PWID are the main group at risk for HCV infection in Spain, and the main group at risk for HIV/HCV coinfection.[3]
- As such, the elimination of HCV among PLWH in Spain requires a focus on elimination of HCV transmission among PWID.
- To our knowledge no published study has explored how to achieve elimination within HIV-infected PWID, and the impact of treatment scale-up to coinfected PWID on the broader HCV epidemic among PWID.

AIM

To assess the population impact of HCV treatment scale-up to HIV+ individuals, and its implications for elimination among HIV+ PWID in Andalusia, Spain, using dynamic modeling.

Methods

A deterministic epidemic coinfection model was developed to simulate the transmission of HIV and HCV due to injecting (HIV and HCV) and sexual risk (only HIV) among PWID, parameterized to data on HCV treatment among HIV-infected individuals in Andalusia [HERACULES cohort[1]].

Model structure: Model schematics are in **Figure 1**. The model incorporates:

- Infection with HIV (uninfected, HIV-infected undiagnosed, HIV-infected diagnosed), HCV (uninfected, HCV chronically infected, HCV chronically infected treatment failure), and HIV/HCV coinfection (stratified by the above stages). The model is additionally stratified by history of injection drug use (denoted by subscript j) with recently initiated PWID (<10 years ago, j=1), non-recently initiated PWID (>10 years ago, j=2) and ex-PWID (j=3). We assume HCV transmission only occurs among PWID, and not ex-PWID.

Model calibration to epidemiological data:

- 45% [range 40-50%] and 60% [range 55-65%] chronic HCV prevalence among PWID who initiated injecting <10 years ago and >10 years ago, respectively, 2010[3].
- 20% [range10-30%] and 40% [range 30-50%] HIV prevalence among PWID who initiated injecting <10 years ago and >10 years ago, respectively, in 2010[3].
- 54% chronic HCV prevalence among HIV+ ever PWID (PWID+ex-PWID) in 2015[4].
- 2,669 [range 2402-2936] HIV+/HCV+ diagnosed ever PWID in 2015. [HERACULES cohort[1]]
- Proportion of HIV+/HCV+ diagnosed individuals who are current PWID in 2015: 14% (range 10-18%). [HERACULES cohort[1]]

We used Bayesian methods for model parameterization and calibration to generate multiple model fits. We sample most parameters from underlying distributions 500 times, calibrating to the epidemiological data using least squares methods, and selecting fits which lay within the range of the HCV and HIV prevalence among PWID data. This resulted in 100 selected model fits.

We implemented an increase in HCV treatment rates to 33%/year among diagnosed coinfected individuals from 2015 onwards (consistent with observations in the HERACULES cohort). We also implemented a halving in the entry rate of new injectors in 2010, consistent with data on entry to opiate substitution therapy in Andalusia.

We model the impact on HCV incidence and chronic prevalence until 2030 with the following scenarios:

1. **Status quo:** Continuing current treatment rates (33%/year among diagnosed coinfected individuals from 2015)
2. **Coinfected PWID scale-up from 2018:** 100% screening and treatment of coinfected PWID
3. **All PWID scale-up from 2018:** As in (2) plus HCV treatment of HIV-negative PWID of 10%/yr
4. **No scale-up from 2015** (counterfactual)

Figure 1. (A) Model Schematic showing HIV and HCV stages (B) Stratification by history of IDU

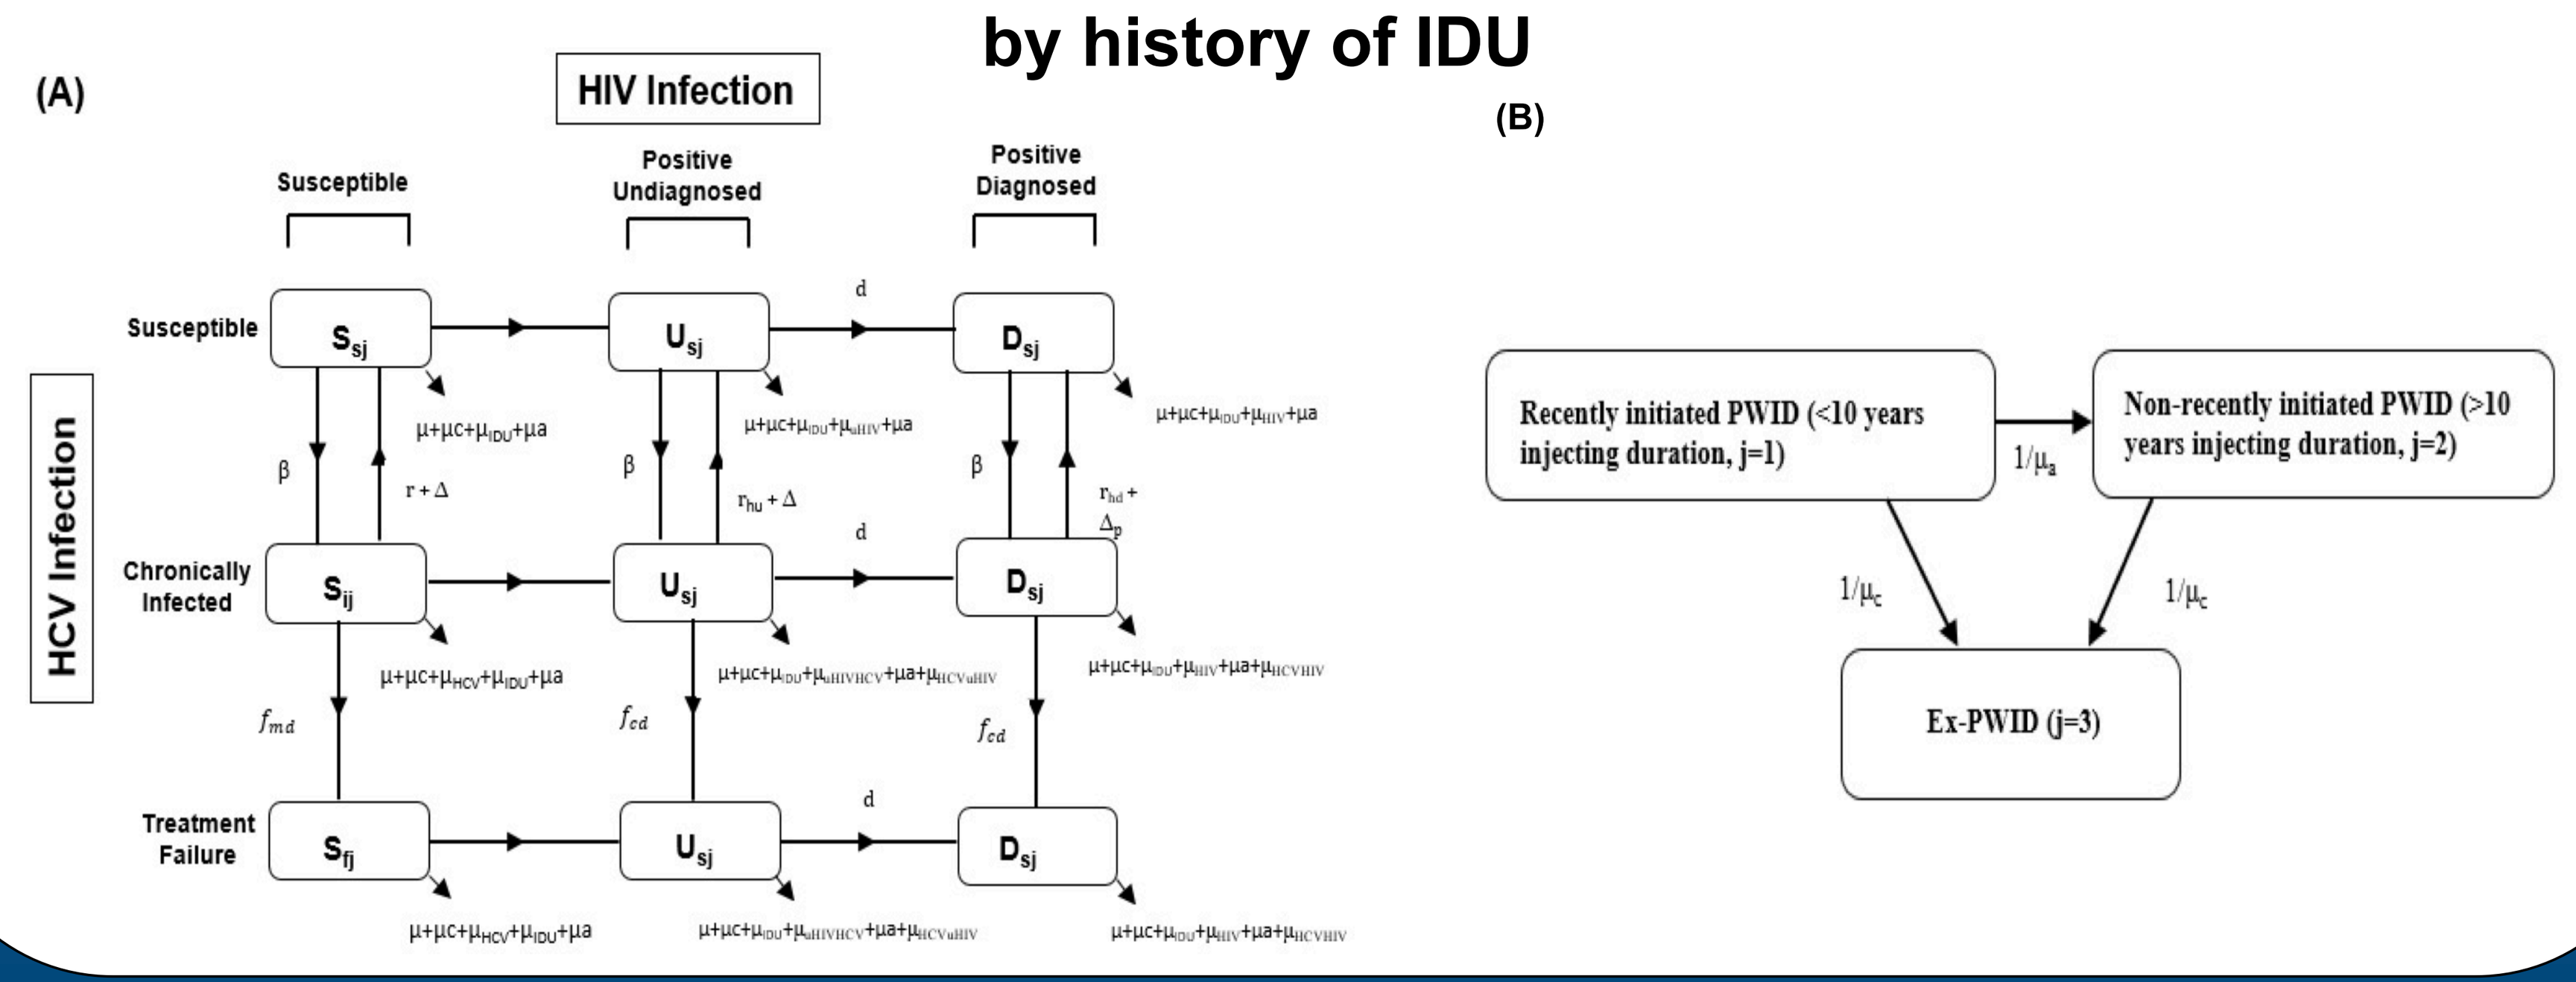
(A)
HIV Infection
Susceptible
Positive Undiagnosed
Positive Diagnosed
Susceptible
Chronically Infected
Treatment Failure
HCV Infection
(B)
Recently initiated PWID (<10 years injecting duration, j=1)
Non-recently initiated PWID (>10 years injecting duration, j=2)
Ex-PWID (j=3)

Results

Epidemic prior to DAA scale-up in 2015

- Prior to DAA scale-up in 2015, we project HCV chronic prevalence was increasing among PWID in Andalusia due to fewer new PWID, leading to an aging cohort.
- We estimated that in 2015, a mean of 29% (95% Confidence Interval[95%CI] 23-34%) of HCV infected PWID and 33% (95%CI 22-43%) of ex-PWID in Andalusia were HIV/HCV coinfected.

Modeled impact of observed DAA scale-up since 2015

- Observed scale-up of DAAs to HIV-diagnosed individuals since 2015 reduced the number of diagnosed coinfected individuals with history of IDU (mean 2948 in 2015 to 1576 in 2017), and reduced chronic prevalence/incidence among PWID (**Fig 2, black line**)

Modeled future impact of DAA treatment scenarios among HIV/HCV coinfected individuals

- Continuing current DAA treatment rates among HIV-diagnosed individuals could dramatically reduce the number of prevalent diagnosed coinfected individuals with a history of IDU (mean 75% relative reduction from 2015-2030).
- However, this would only reduce HCV chronic prevalence by a mean relative 21% and 9% from 2015 among HIV+ PWID and all PWID, respectively, by 2030. This moderate reduction is likely due to transmission from PWID with HCV monoinfection and undiagnosed coinfection (**Fig 2, black line).**
- If all coinfected PWID were diagnosed and treated annually from 2018, this could lead to a mean 74% relative reduction from 2015 in chronic prevalence among HIV+ PWID by 2030. However, this would only halve HCV incidence by 2030 among HIV+ and all PWID (**Fig 2, blue line**).
- If treatment were scaled-up between HIV+ and HIV- negative PWID, greater impact could be achieved (**Fig 2, green line**)

Figure 2. Mean model projections for HCV chronic prevalence and incidence among people who inject drugs (A,C) and HIV+ people who inject drugs (B,D) in Andalusia, Spain with various treatment scenarios and 90% SVR.

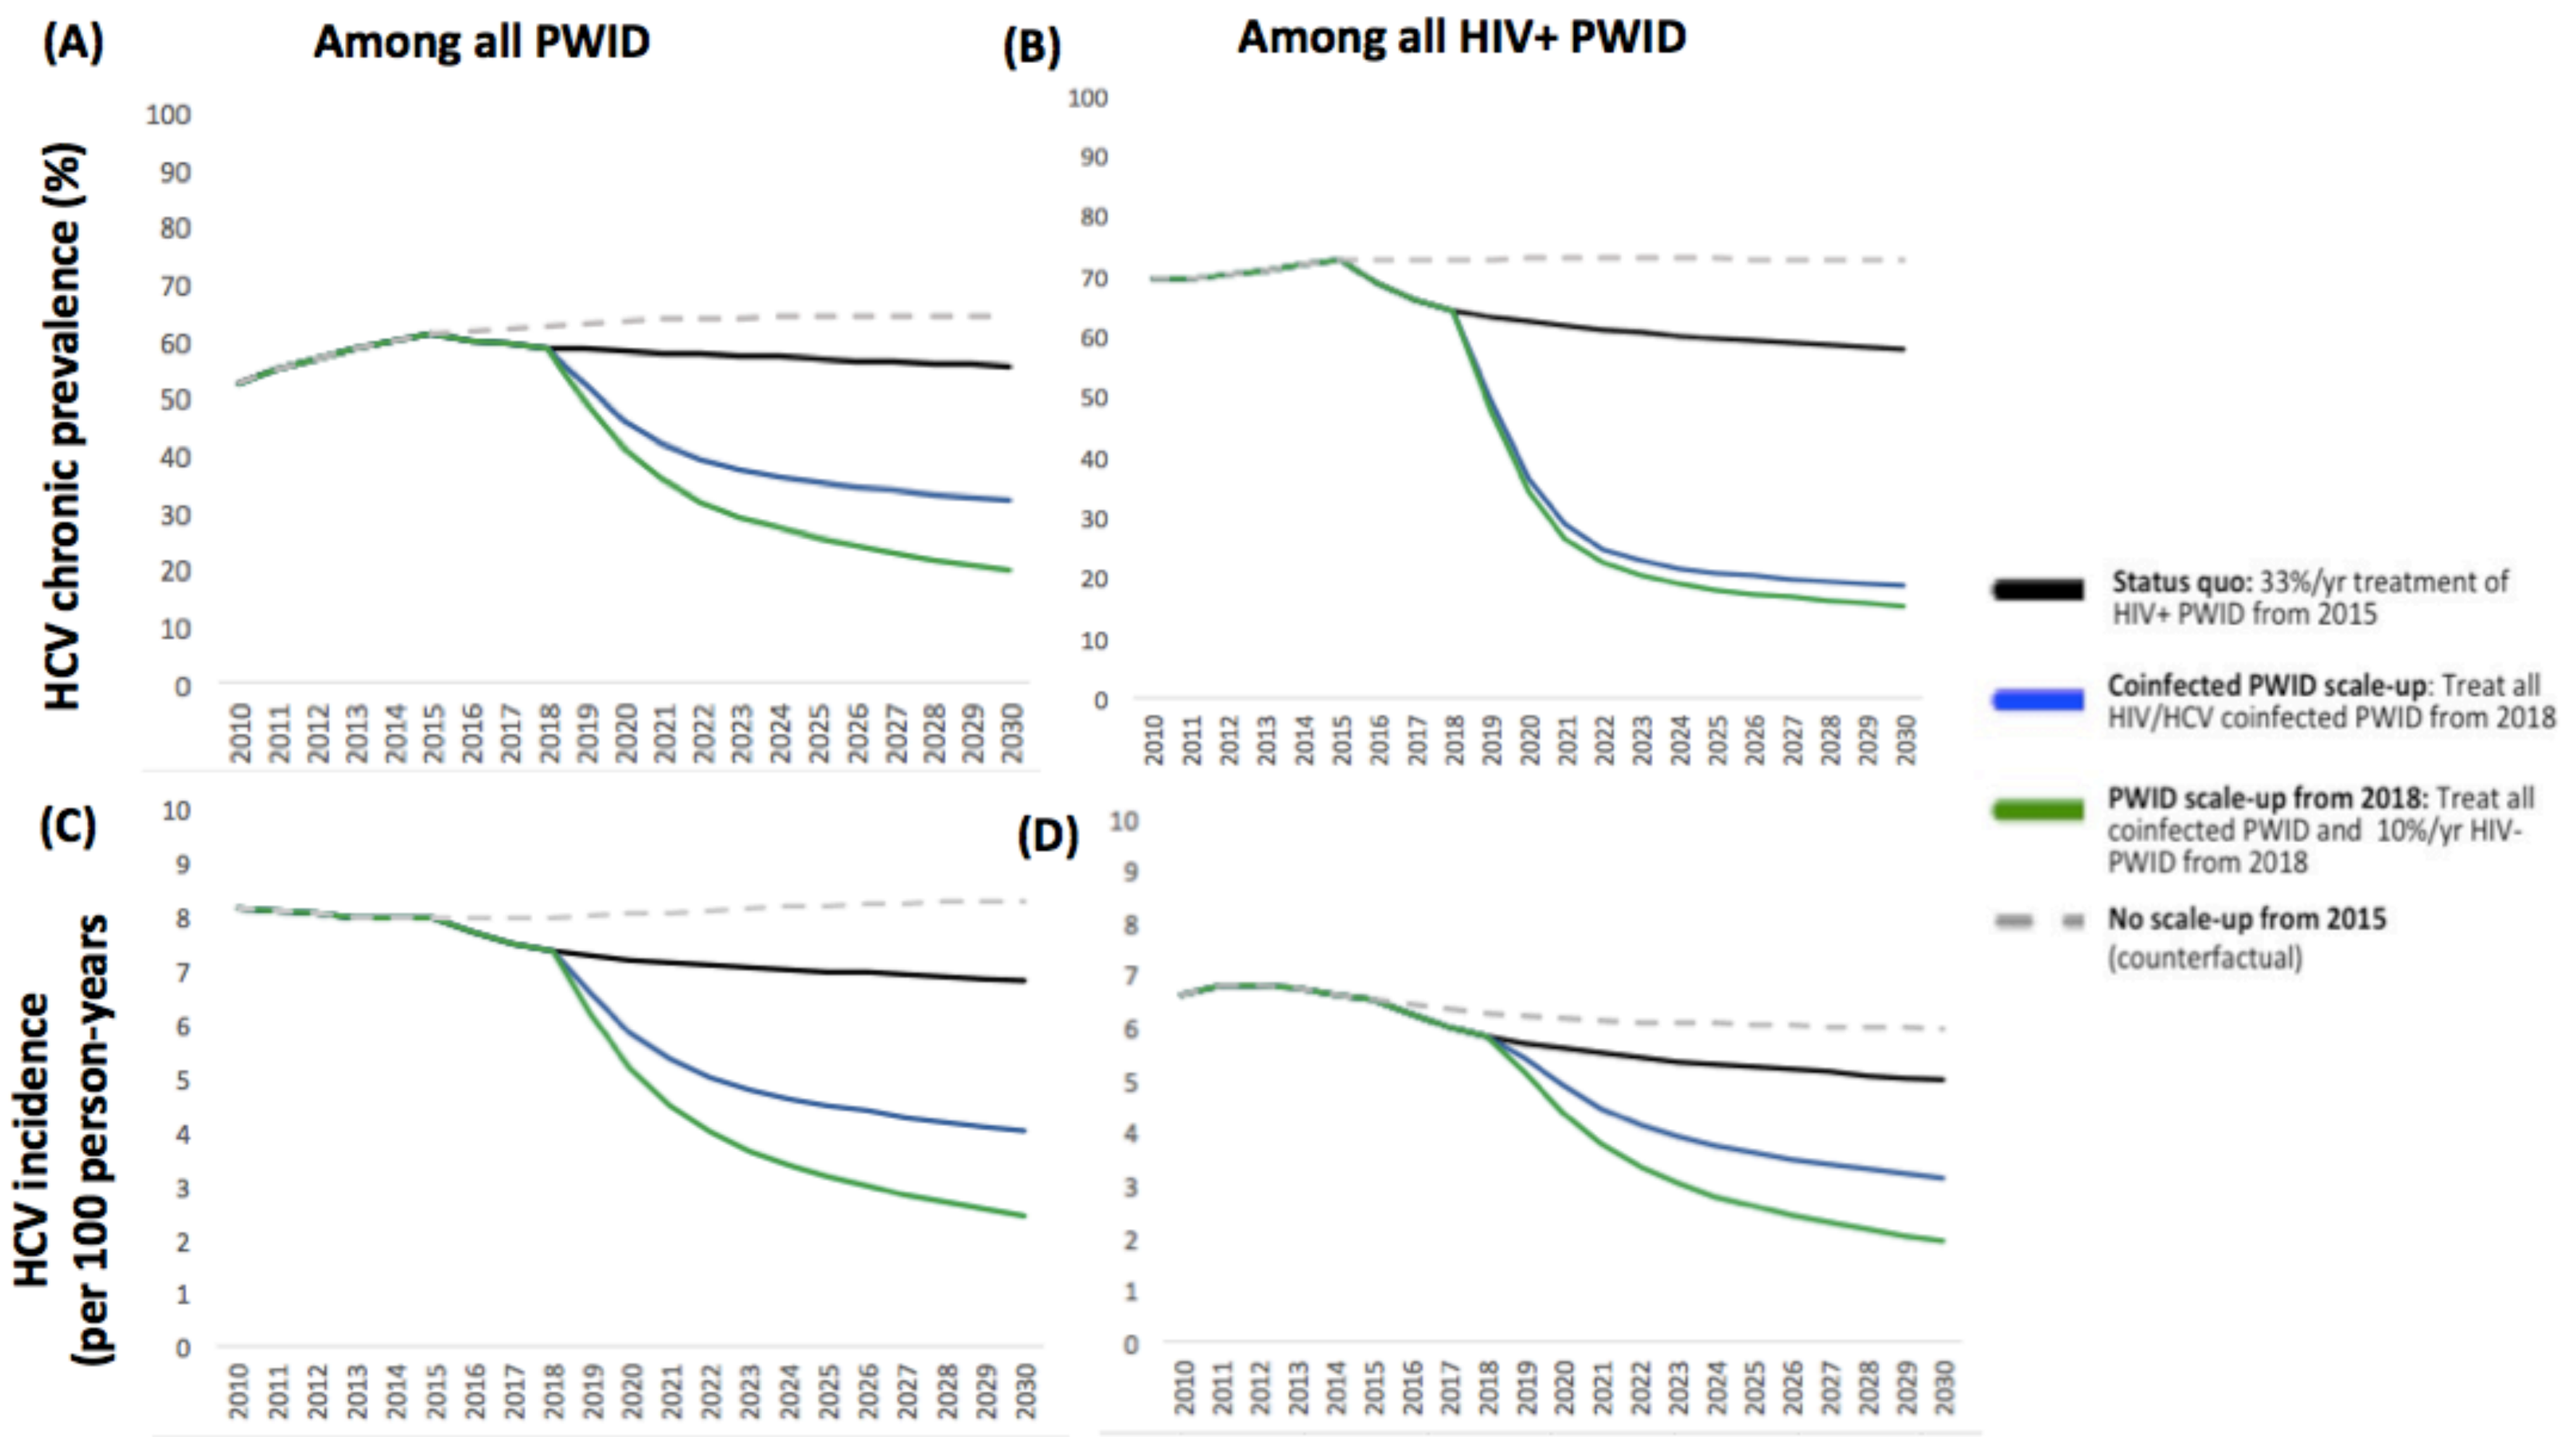
(A) Among all PWID
(B) Among all HIV+ PWID
HCV chronic prevalence (%)
(C)
(D)
HCV incidence (per 100 person-years)
Status quo: 33%/yr treatment of HIV+ PWID from 2015
Coinfected PWID scale-up: Treat all HIV/HCV coinfected PWID from 2018
PWID scale-up from 2018: Treat all coinfected PWID and 10%/yr HIV- PWID from 2018
No scale-up from 2015 (counterfactual)

Conclusions

- Recent scale-up of HCV treatment to HIV/HCV coinfected individuals in Spain since 2015 may have had an impact on reducing HCV incidence and chronic prevalence among the broader PWID population in Andalusia.
- Further scale-up to diagnose and treat all HIV+ PWID each year could halve HCV incidence among PWID by 2030, but will likely not achieve the WHO elimination goal of 90% reduction in HCV incidence by 2030.
- Elimination efforts should focus on HCV diagnosis and treatment among both coinfected and monoinfected PWID.

References

1. Rivero-Juarez A, et al. Dimension of chronic hepatitis C virus in HIV-infected patients in the interferon-free era: an overview from south Spain. *Eur J Clin Microbiol Infect Dis* 2015 Nov; 34(11):2247-55..
2. WHO Global health sector strategy on viral hepatitis 2016-2021. 2017. Available from: http://apps.who.int/iris/bitstream/10665/246177/1/WHO-HIV-2016.06-eng.pdf?ua=1
3. Folch C, et al. High Prevalence and Incidence of HIV and HCV Among New Injecting Drug Users With a Large Proportion of Migrants--Is Prevention Failing? *Subst Use Misuse* 2016 28;51(2):250-60.
4. Berenguer J, et al. Human immunodeficiency Virus/Hepatitis C Virus Coinfection in Spain: Prevalence and Patient Characteristics. *Open Forum Infec Dis* 2016; 2(3):ofw059.

Acknowledgements

This study was funded by Gilead Sciences. The funder had no influence on the design, analysis and content of the study. NM has received grants and honoraria from Gilead and Merck.

Contact Information

Britt Skaathun, MPH, PhD
bskaathun@ucsd.edu